Oral Long-Acting Menin Inhibitor, BMF-219, Normalizes Type 2 Diabetes Mellitus in Two Rat Models

We Aim to Cure™

Thomas Butler, MSc MBA[1], Weiqun Li, PhD, Brian Law, BS [1], Tenley Archer, PhD,, Taisei Kinoshita, PhD[1], Mini Balakrishnan, PhD[1] and Priyanka Somanath, PhD[1]

[1]Biomea Fusion, Inc. Redwood City, CA

Introduction

- Menin is a scaffold protein, encoded by the gene *MEN1*, that has been recently recognized for its role in Type 2 Diabetes Mellitus (T2DM) as a key regulator of β-cell proliferation.
- *Men1* knockout mice demonstrate increased B-cell mass generation (Yang et al., 2010)
- Menin inhibition has previously been shown to improve glycemic control in high fat induced diabetic mice (Ma et al., 2021).
- Men1-excised mice do not develop hyperglycemia in a Streptozotocin-(STZ) induced rat model, which is a model for impaired beta-cell function and insulin production, demonstrating the role of menin in glycemic control.
- BMF-219 is an orally bioavailable, selective, covalent menin inhibitor that elicits a broad impact on the complexes surrounding menin that also direct its biological function.
- Herein, we report the first evidence that BMF-219 restores glycemic control in Zucker Diabetic Fatty (ZDF) Rat and Streptozotocin-induced Rat (STZ) models of T2DM, with prolonged glycemic control two weeks after dosing in ZDF rats.

Methods

ZDF rats were treated daily with BMF-219, vehicle, or pioglitazone for 16 days and monitored for an additional two weeks post-treatment, until Day 29. STZ rat models were induced through pre-treatment of animals on a high-fat diet with low doses of streptozotocin at day -14 and day -7 prior to starting treatment at Day 0 until Day 16. ZDF and STZ rats were monitored for fasting blood glucose levels, oral glucose tolerance test (OGTT), insulin and C-peptide levels, HOMA-IR (Homeostatic Model Assessment of Insulin Resistance), blood lipemic levels and body weight according to the schematic for each model below. ZDF rats were analyzed for indicated readouts for 15 days post-treatment for OGTT at day 29, and serum insulin and C-peptide at day 31.

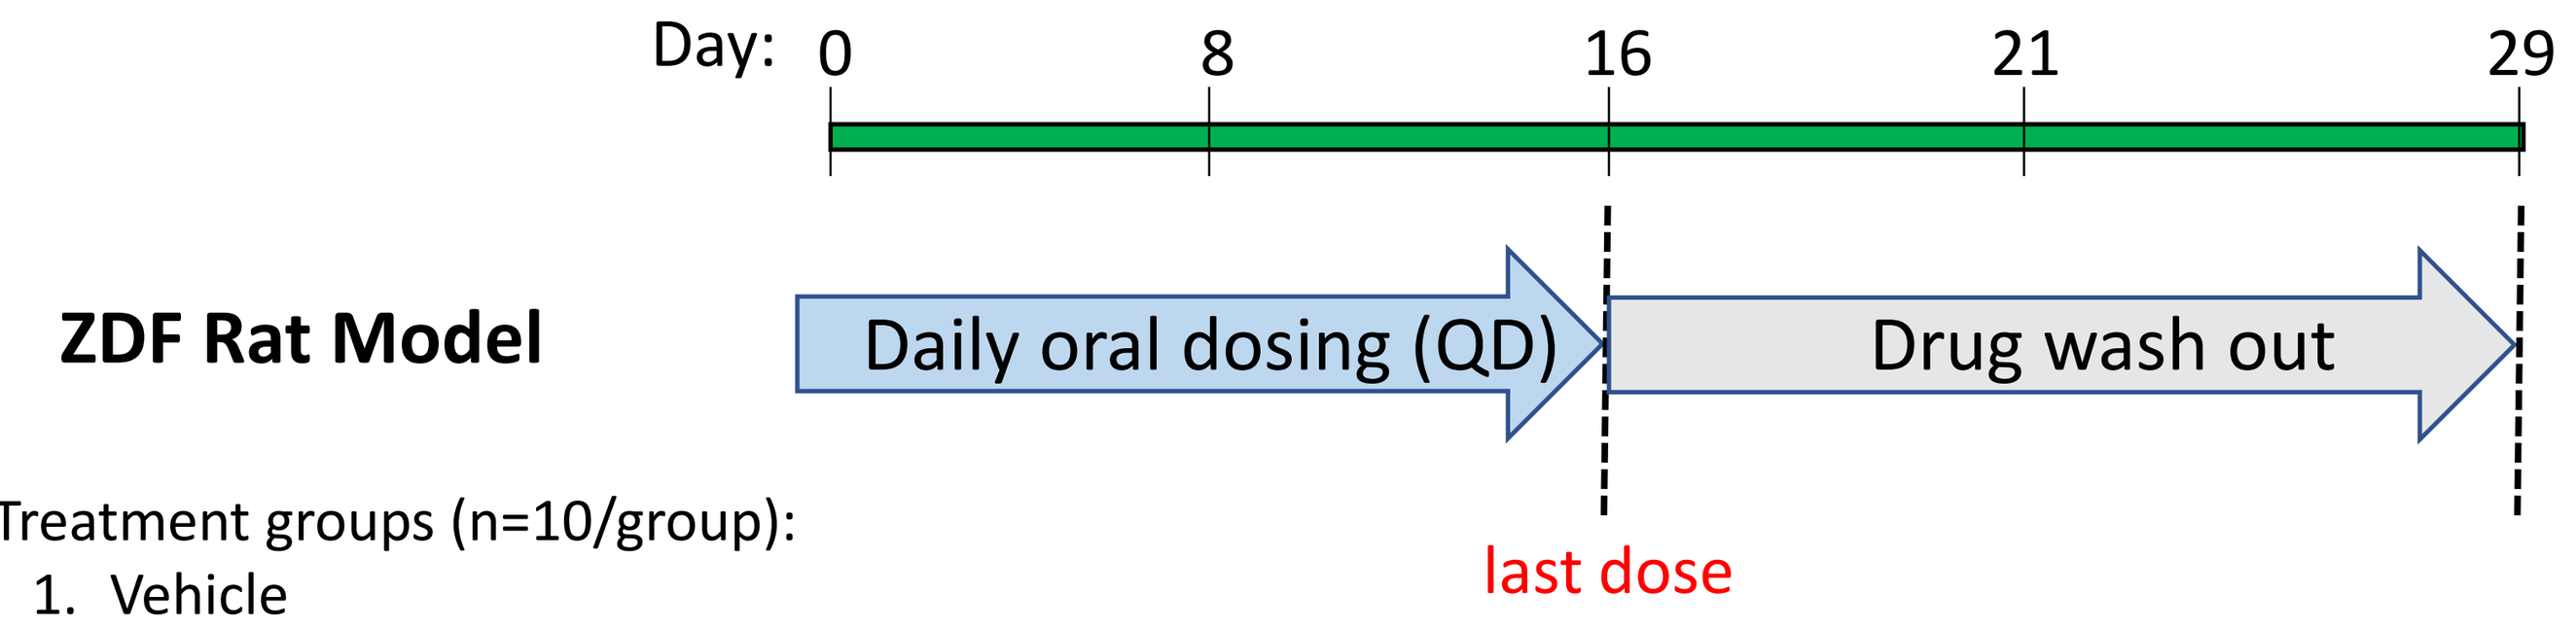
Day: 0 8 16 21 29
ZDF Rat Model
Daily oral dosing (QD)
Drug wash out
last dose

Treatment groups (n=10/group):
1. Vehicle
2. BMF-219 175 mg/kg
3. Pioglitazone 30 mg/kg

Rats monitored for the following parameters through dosing and washout phases include:
Body weight, fasting blood glucose, blood insulin, C-peptide, and OGTT

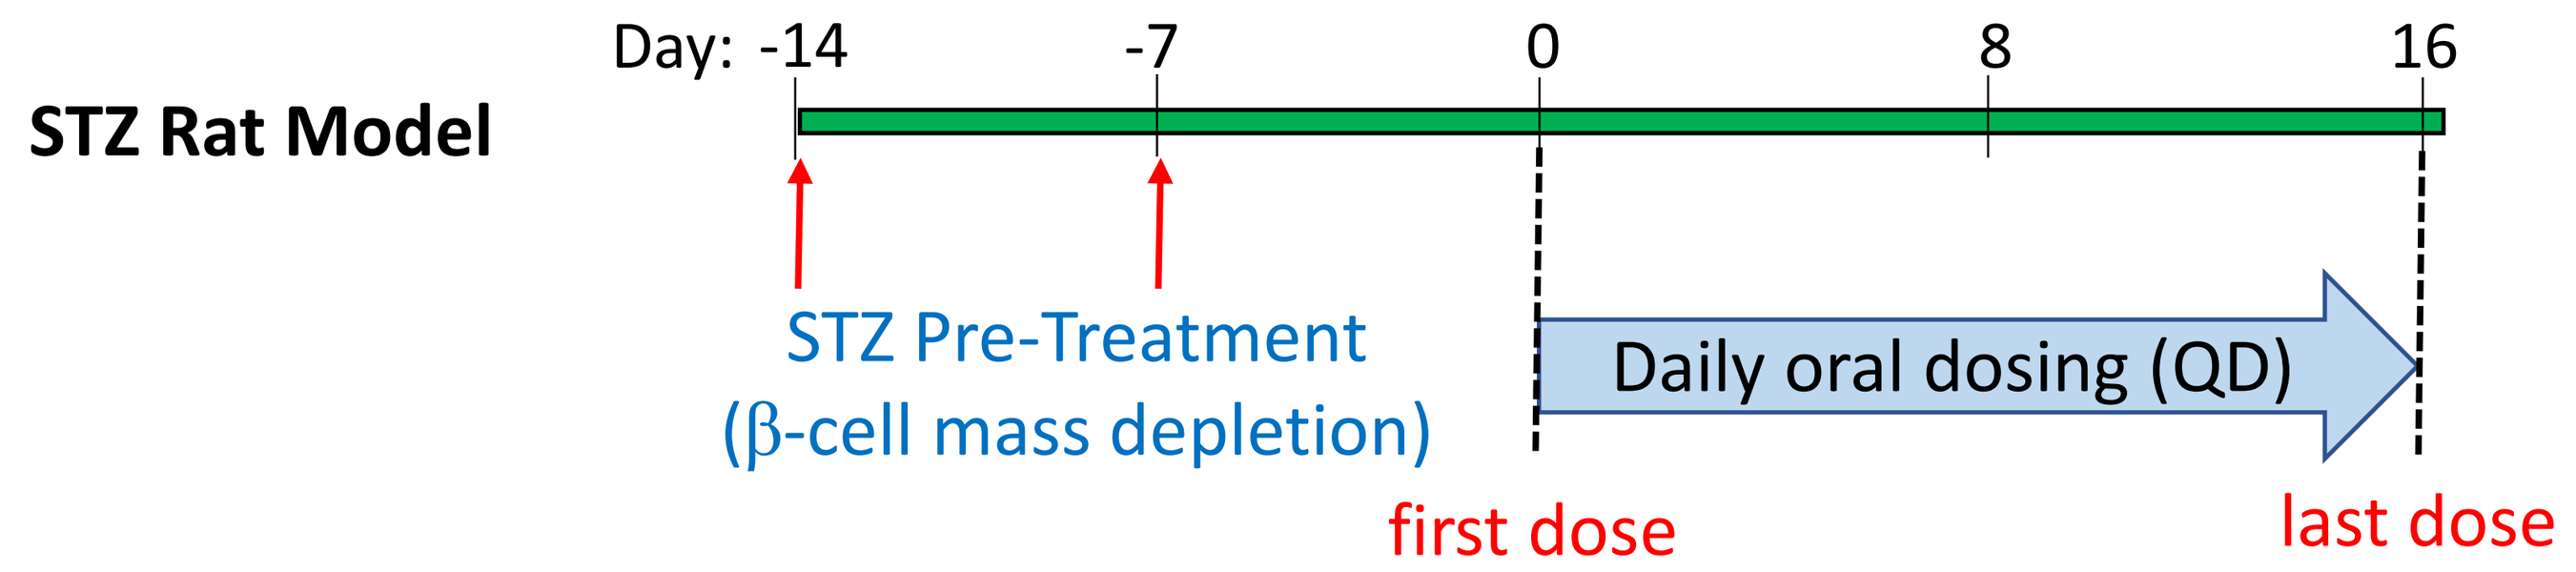
Day: -14 -7 0 8 16
STZ Rat Model
STZ Pre-Treatment
(β-cell mass depletion)
Daily oral dosing (QD)
first dose
last dose

Treatment groups (n=10/group):
1. Vehicle
2. BMF-219 175 mg/kg
3. Pioglitazone 30 mg/kg

Rats monitored for the following parameters through dosing include: OGTT, blood glucose levels

BMF-219 demonstrates significant glycemic control in ZDF Rats

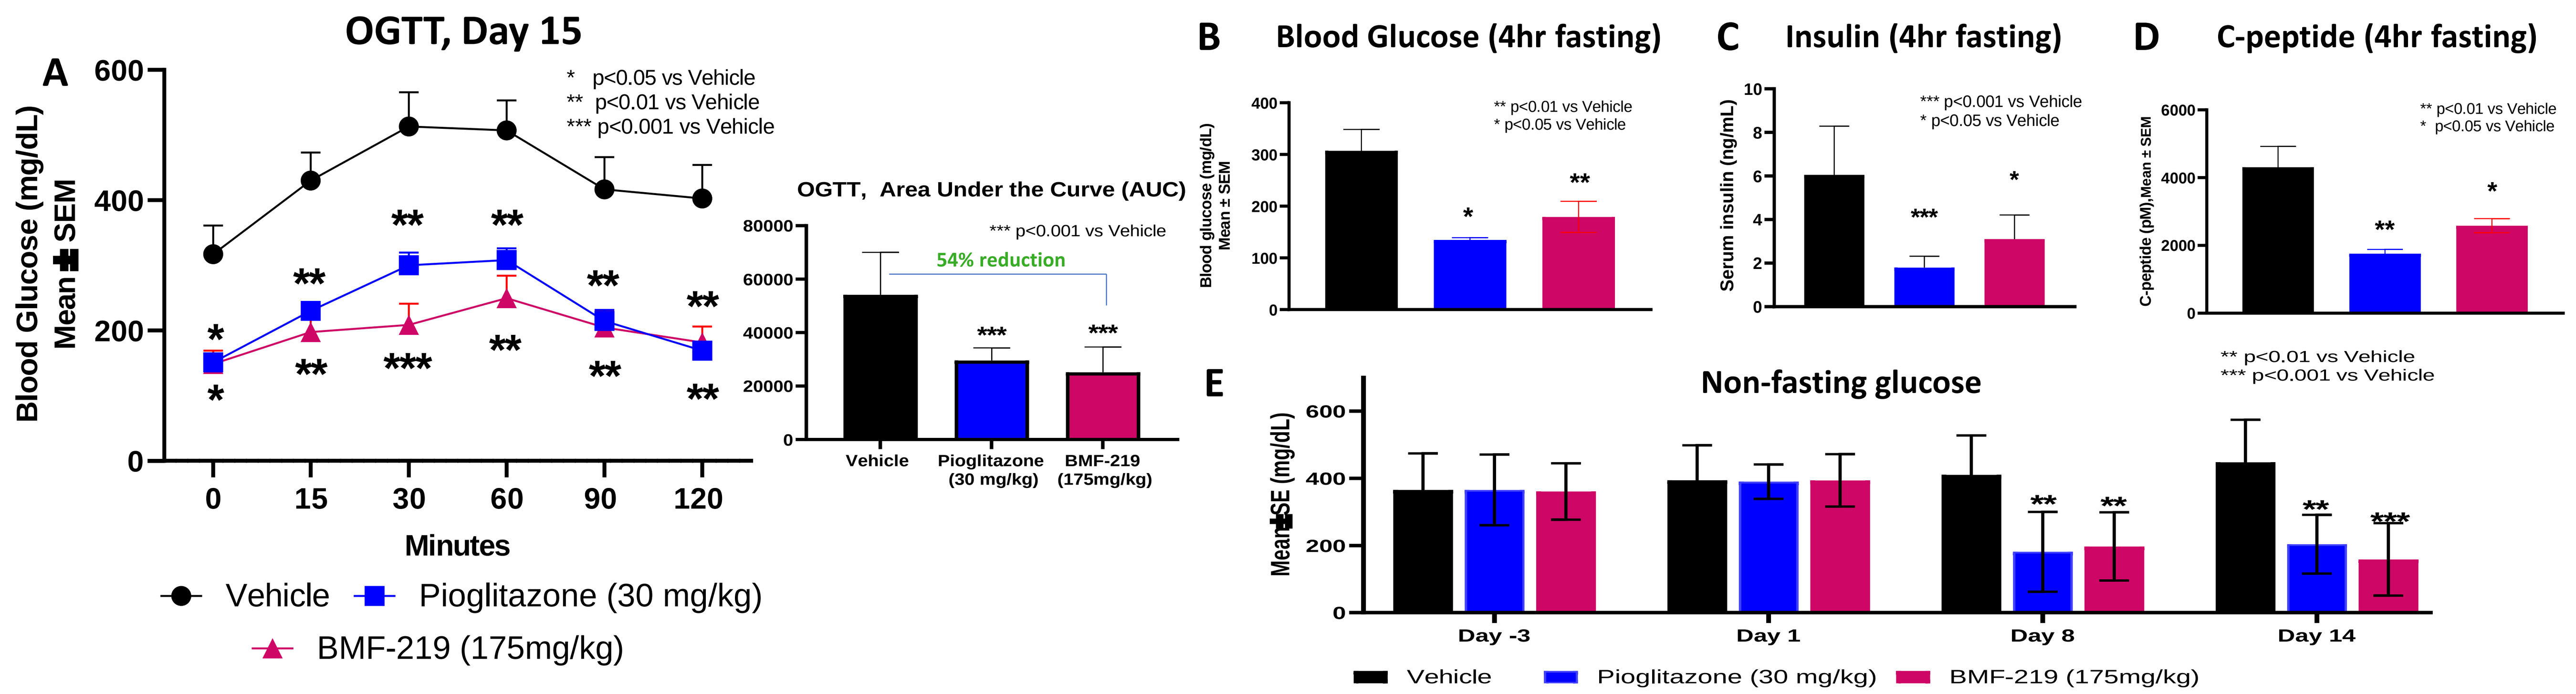
A
OGTT, Day 15
* p<0.05 vs Vehicle
** p<0.01 vs Vehicle
*** p<0.001 vs Vehicle
Blood Glucose (mg/dL) Mean ± SEM
Minutes
Vehicle
Pioglitazone (30 mg/kg)
BMF-219 (175mg/kg)
OGTT, Area Under the Curve (AUC)
*** p<0.001 vs Vehicle
54% reduction
B
Blood Glucose (4hr fasting)
C
Insulin (4hr fasting)
D
C-peptide (4hr fasting)
E
Non-fasting glucose
Day -3
Day 1
Day 8
Day 14

Figure 1. BMF-219 significantly reduces blood glucose levels and alters serum insulin and C-peptide levels in ZDF rats. ZDF rats treated with BMF-219, pioglitazone or vehicle for 16 days were evaluated at various time points. Day 15 OGTT results are shown as time course and AUC (A). Day 17 fasting blood glucose levels (B), fasting insulin (C), and fasting C-peptide (D) are shown. Non-fasting blood sugar was measured weekly on Days -3, 1, 8 and 14 (E). Statistical significance was calculated for treatment groups in comparison with vehicle control.

BMF-219 displays strong glycemic control two weeks after last dose in ZDF Rats

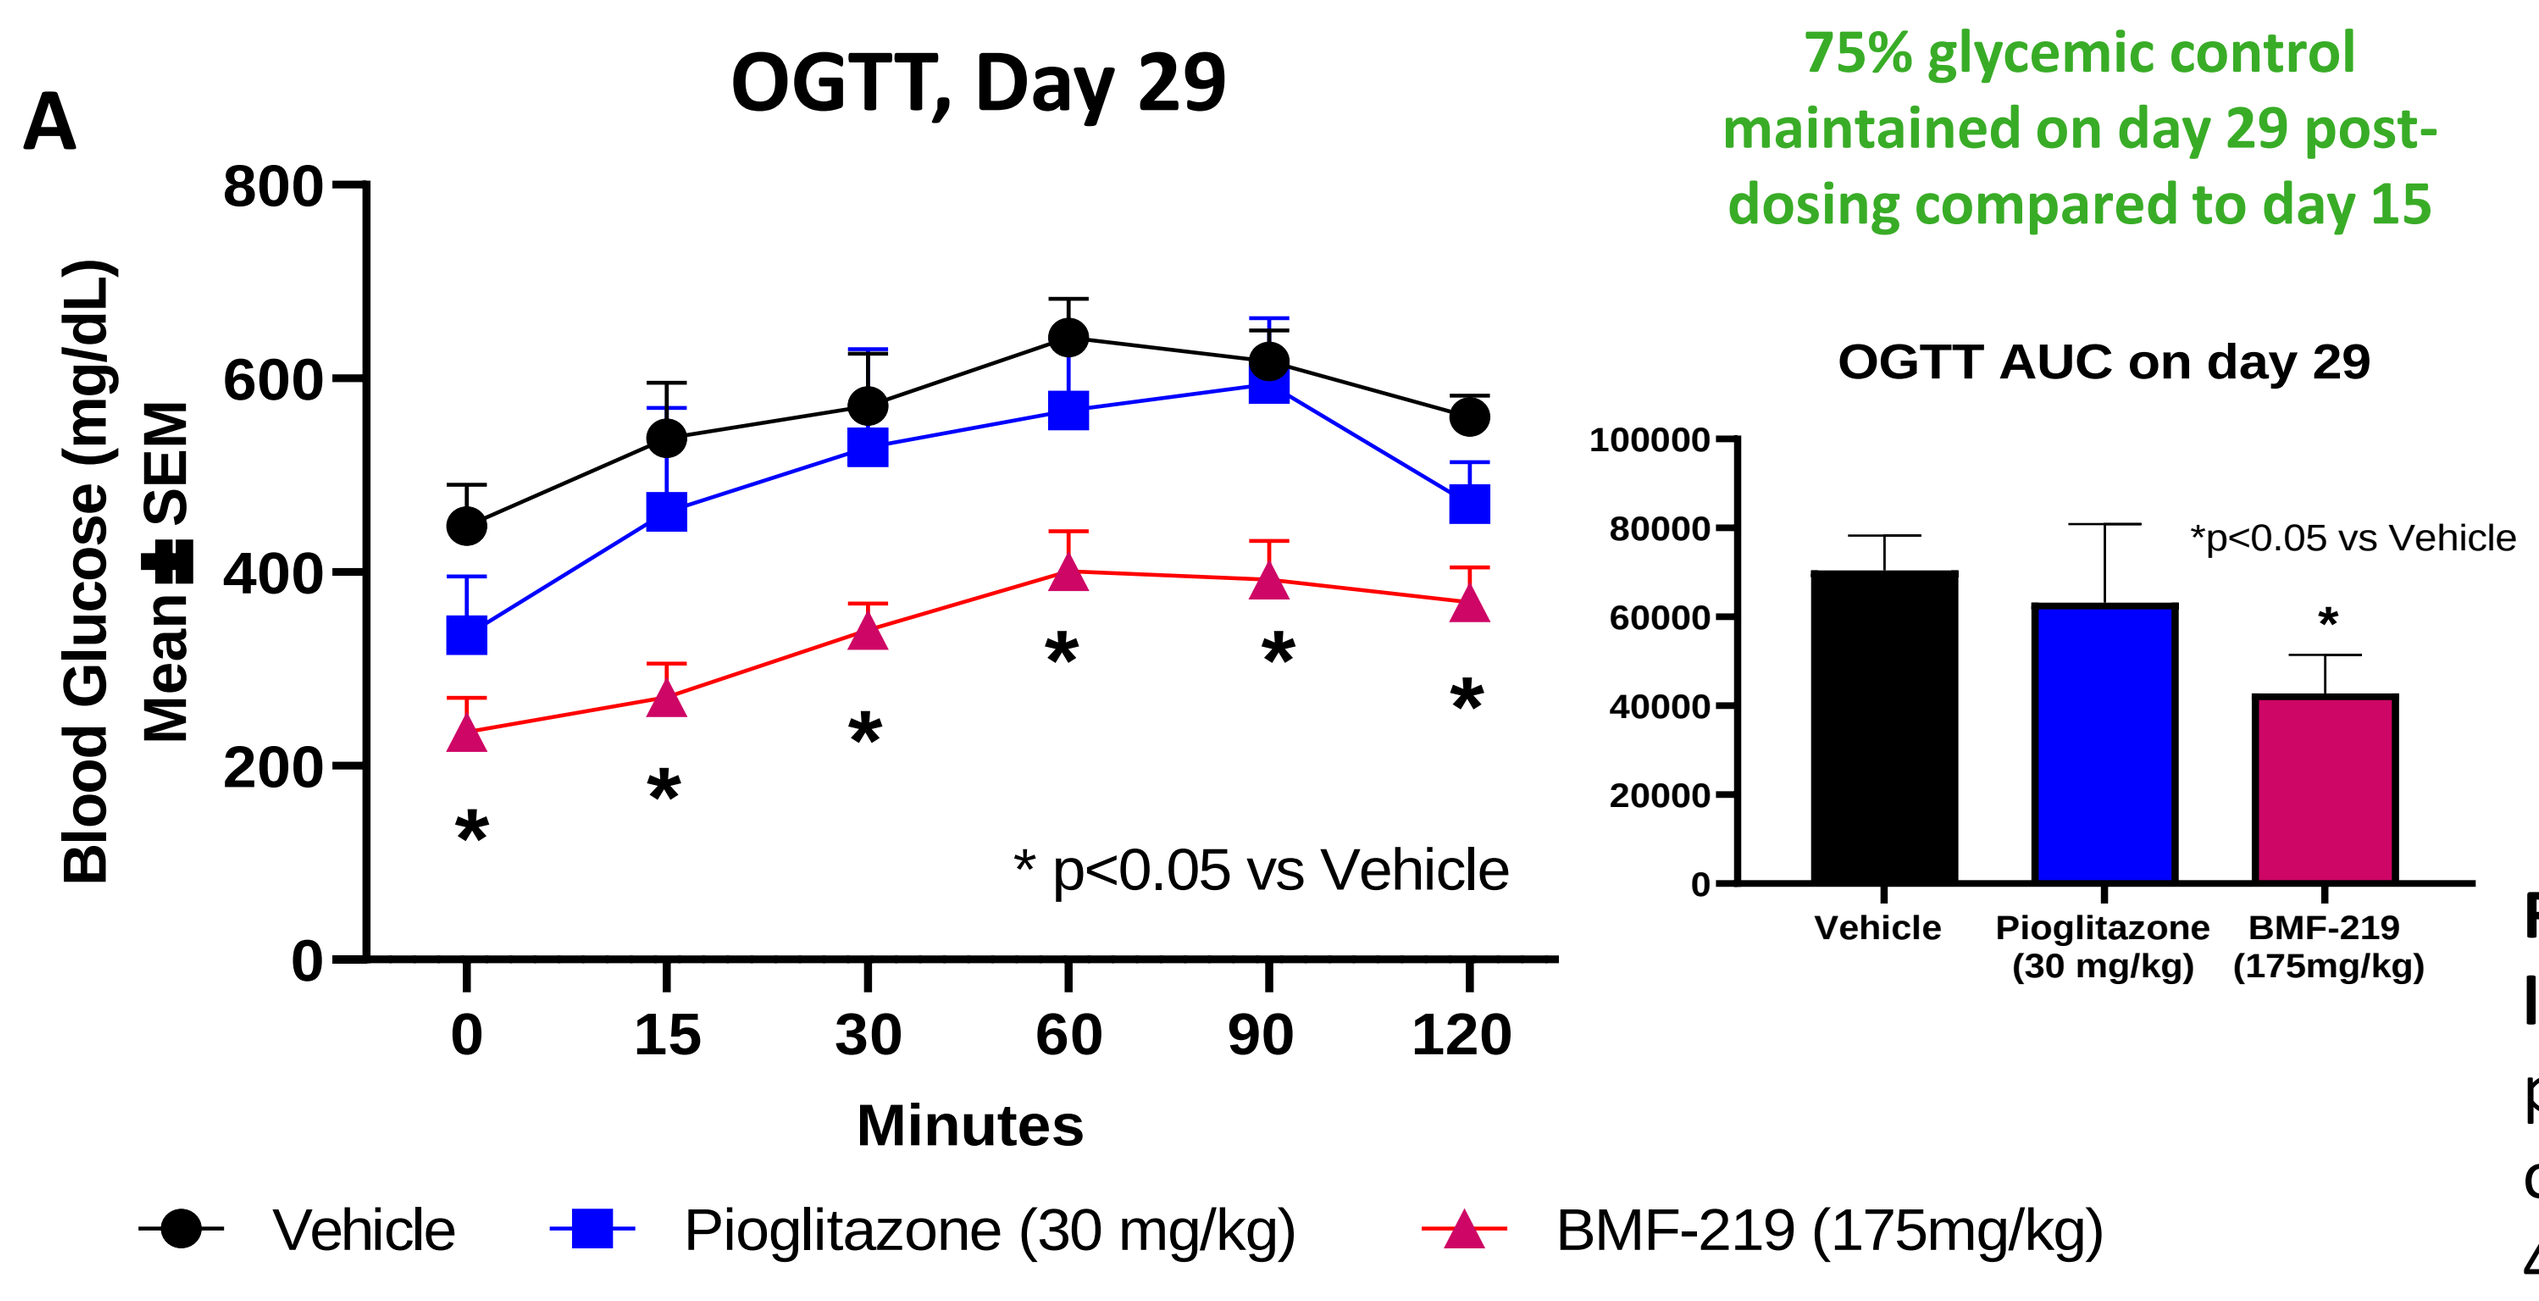
A
OGTT, Day 29
75% glycemic control maintained on day 29 post-dosing compared to day 15
OGTT AUC on day 29
*p<0.05 vs Vehicle
Blood Glucose (mg/dL) Mean ± SEM
Minutes
Vehicle
Pioglitazone (30 mg/kg)
BMF-219 (175mg/kg)

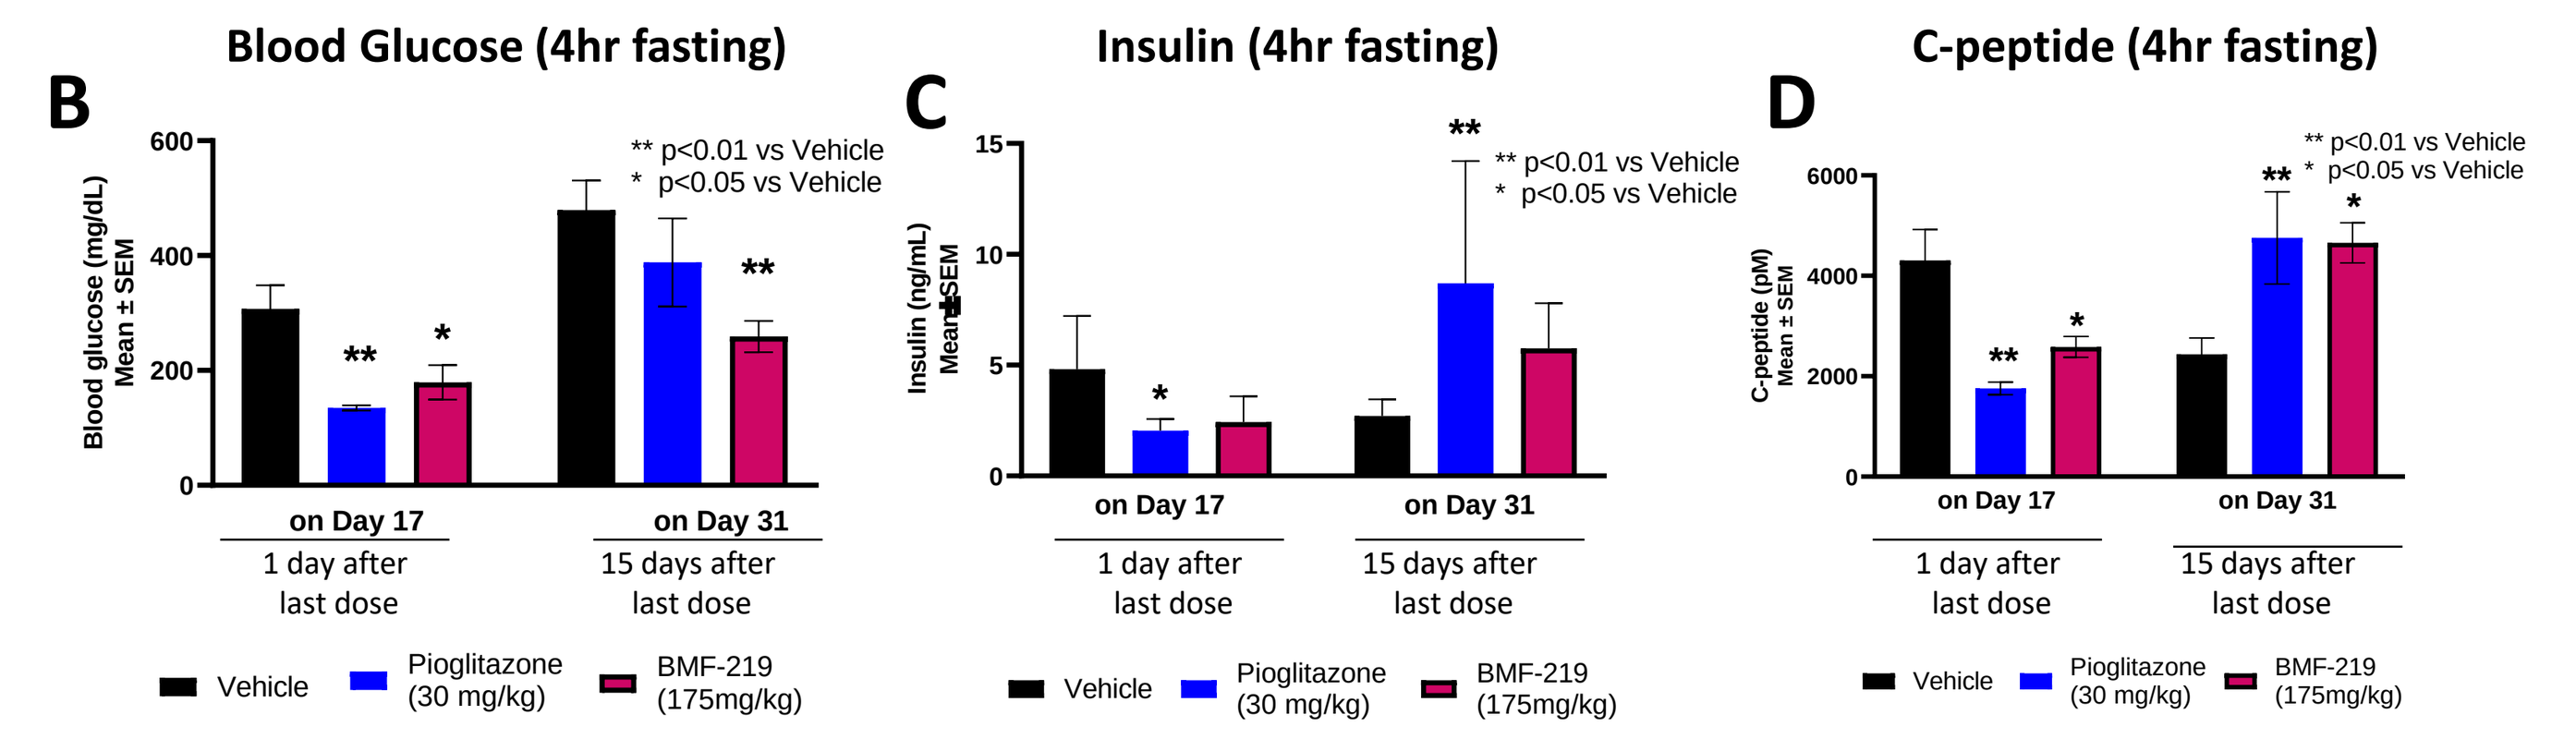
B
Blood Glucose (4hr fasting)
C
Insulin (4hr fasting)
D
C-peptide (4hr fasting)
on Day 17
1 day after last dose
on Day 31
15 days after last dose

Figure 2. BMF-219 maintains significant impact on blood glucose, insulin and C-peptide levels during drug washout (two weeks after last dose). ZDF rats treated with BMF-219, pioglitazone or vehicle control for 16 days were monitored for blood glucose levels by OGTT on day 29, ~2 weeks after administration of the last dose, displaying an AUC reduction of 40%, (p<0.05) (A), and on day 31 monitored for 4-hour fasting blood glucose (B), fasting serum insulin (C), and fasting C-peptide levels (D). Statistical significance was calculated for treatment groups in comparison to vehicle control.

BMF-219 achieves marked glycemic control in STZ-induced Rats

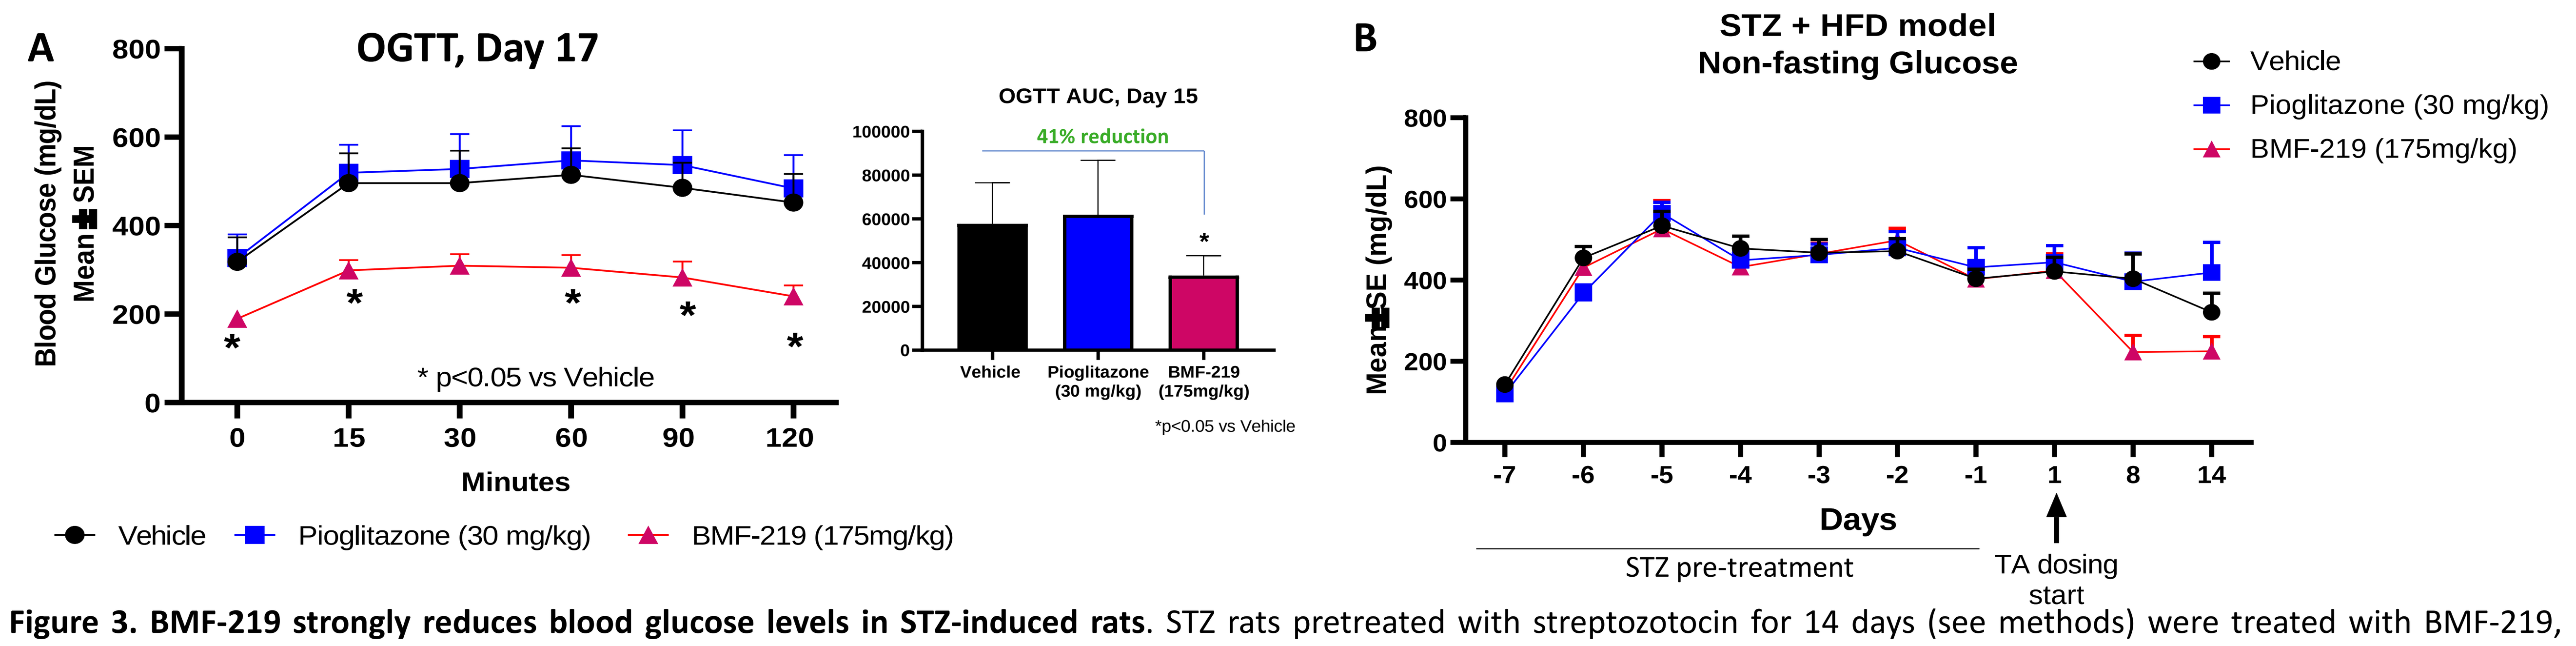
A
OGTT, Day 17
* p<0.05 vs Vehicle
OGTT AUC, Day 15
41% reduction
Minutes
B
STZ + HFD model
Non-fasting Glucose
Days
STZ pre-treatment
TA dosing start
Vehicle
Pioglitazone (30 mg/kg)
BMF-219 (175mg/kg)

Figure 3. BMF-219 strongly reduces blood glucose levels in STZ-induced rats. STZ rats pretreated with streptozotocin for 14 days (see methods) were treated with BMF-219, pioglitazone or vehicle control for 16 days. Blood glucose was measured by OGTT on Day 17 and displayed an AUC reduction of 41%, (p<0.05) in BMF-219 treated rats only with no change to the pioglitazone treated group (A). Non-fasting glucose levels were measured daily during model establishment (STZ treatment) and weekly during treatment with BMF-219, pioglitazone or vehicle control, displaying reduction of glucose levels in BMF-219 treated rats throughout the duration of treatment (B).

BMF-219 increases insulin sensitivity in ZDF rats

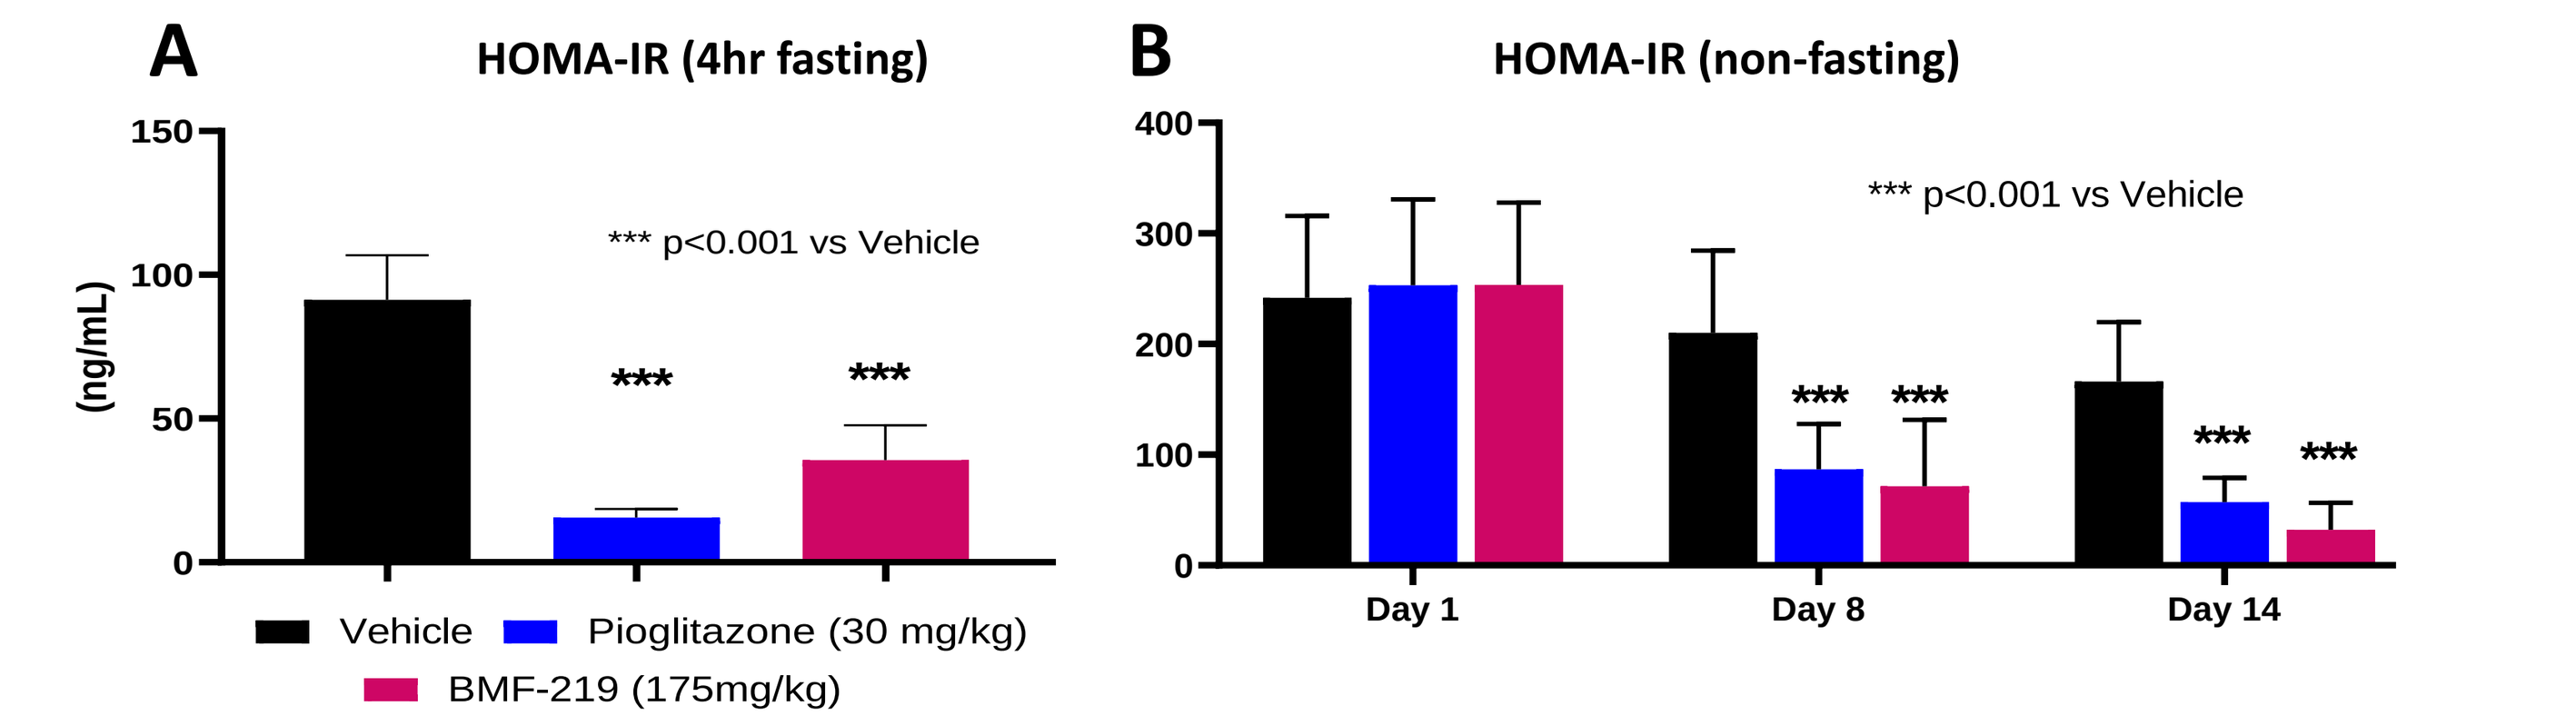
A
HOMA-IR (4hr fasting)
*** p<0.001 vs Vehicle
B
HOMA-IR (non-fasting)
Day 1
Day 8
Day 14
Vehicle
Pioglitazone (30 mg/kg)
BMF-219 (175mg/kg)

Figure 4. Measurement of HOMA-IR in rats treated with BMF-219 for 16 days. ZDF rats treated with BMF-219, pioglitazone or vehicle were analyzed for HOMA-IR fasting at day 17(A) or non-fasting (B) and values were compared to vehicle control to calculate statistical significance.

BMF-219 significantly reduces blood lipemic levels and reduces body weight in treated ZDF rats

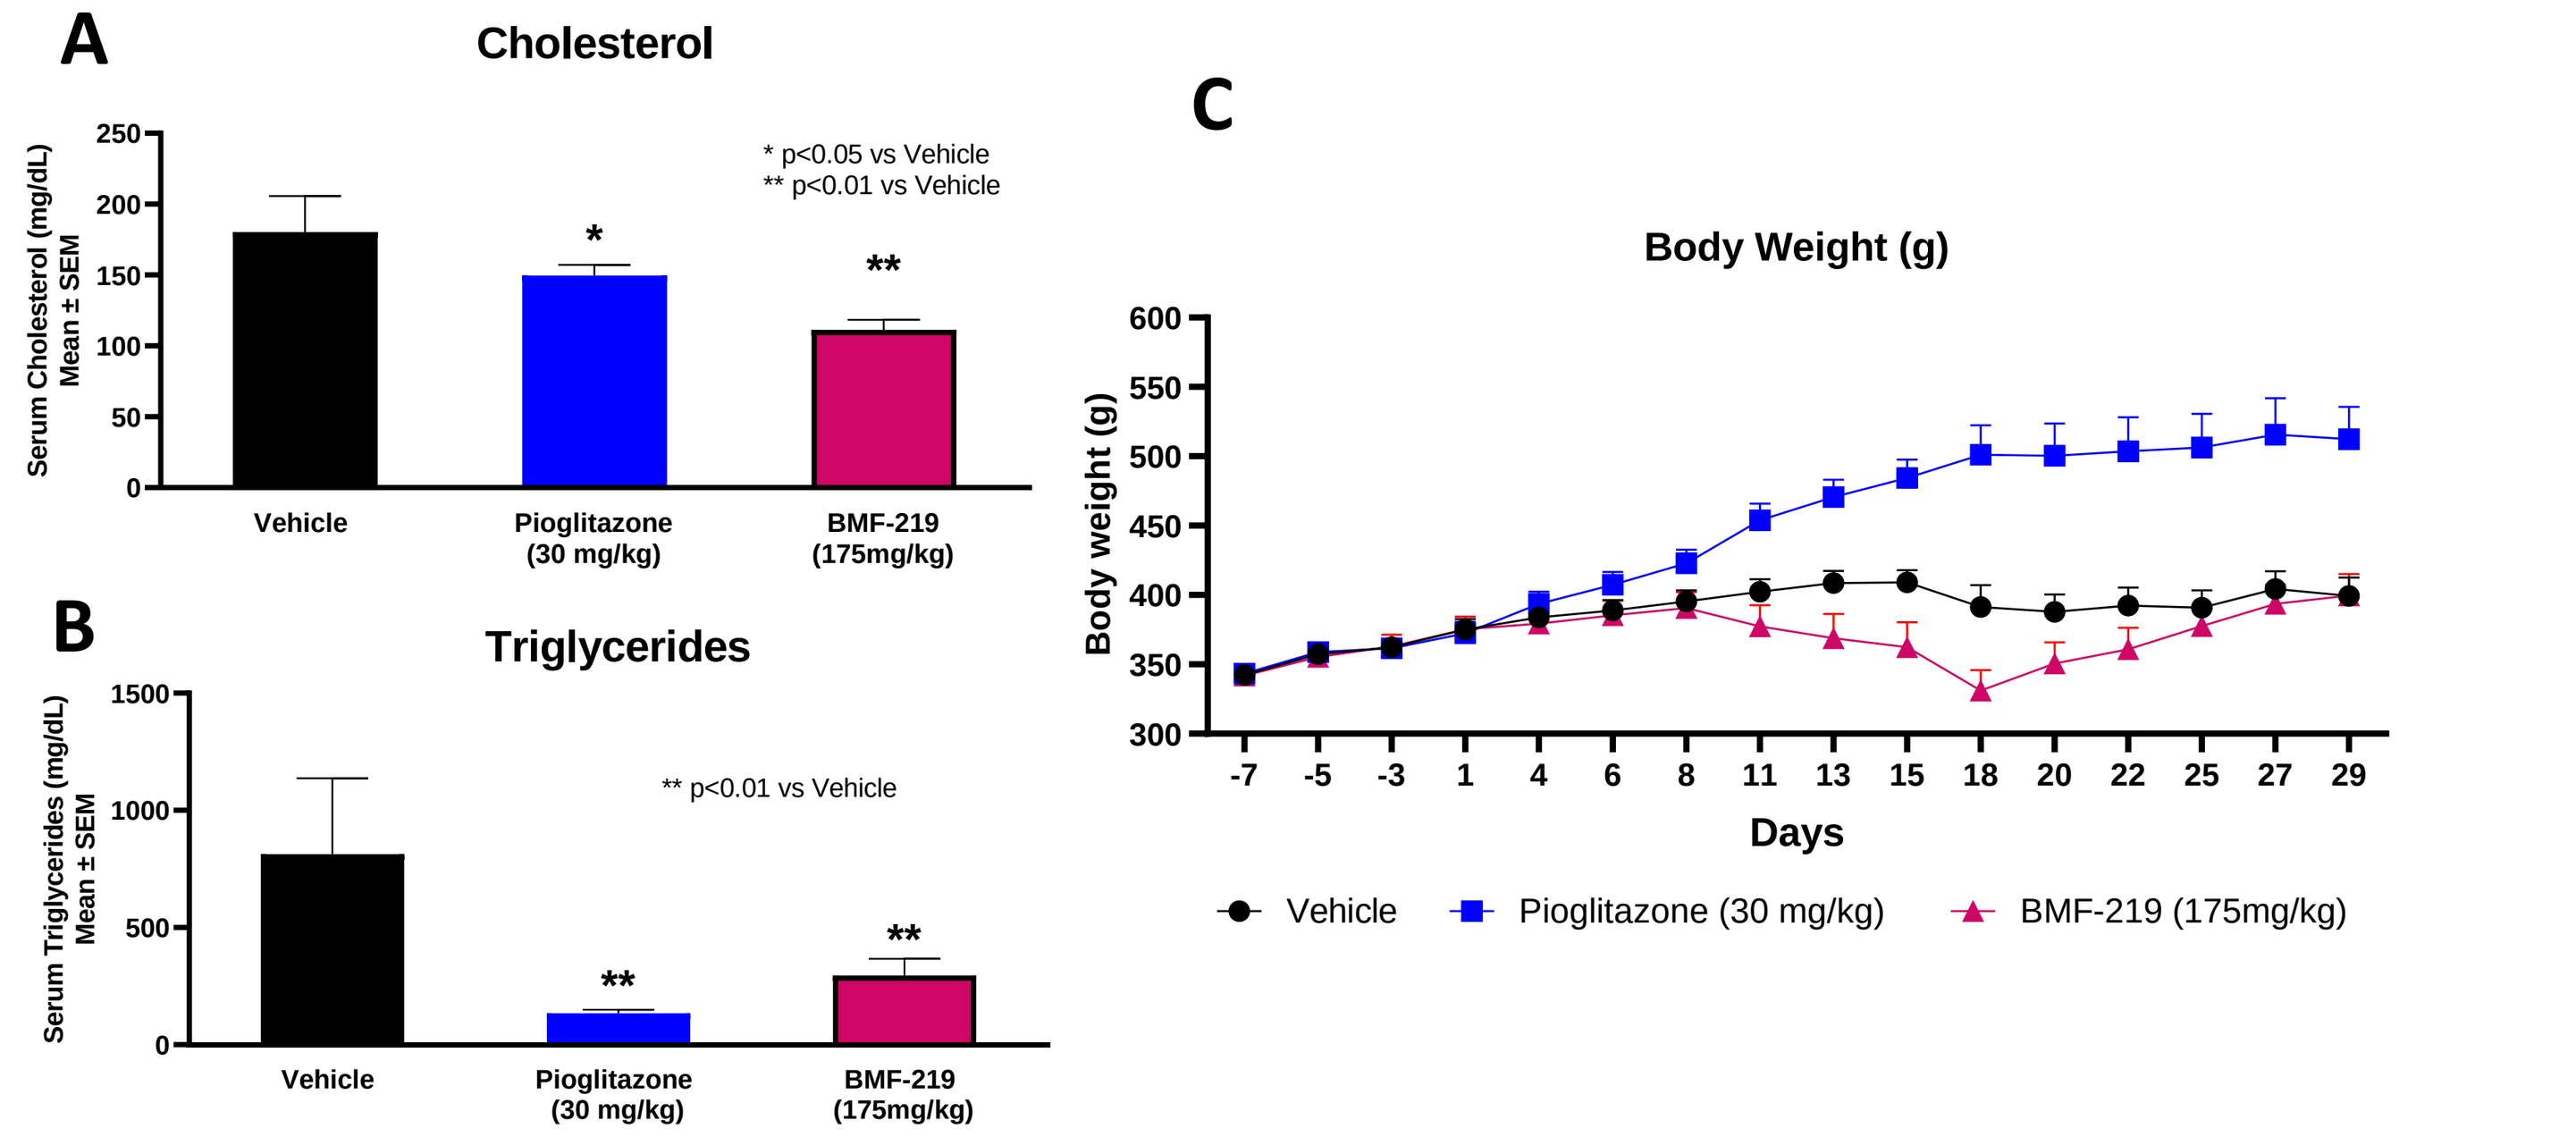
A
Cholesterol
* p<0.05 vs Vehicle
** p<0.01 vs Vehicle
B
Triglycerides
** p<0.01 vs Vehicle
C
Body Weight (g)
Days
Vehicle
Pioglitazone (30 mg/kg)
BMF-219 (175mg/kg)

Figure 5. Measurement of cholesterol, triglycerides and body weight in BMF-219 treated rats for 16 days. Cholesterol (A) and triglycerides (B) were measured at Day 17. Body weight was measured daily during treatment and continually monitored two weeks after treatment (C). All groups were treated with vehicle, BMF-219 or pioglitazone and compared to vehicle for statistical analyses. Animals continued to eat a high fat diet until Day 29.

Conclusions

- BMF-219 was tested in rats at clinically relevant exposures.
- BMF-219 treatment resulted in a significant reduction (~50%) in fasting and non-fasting blood glucose levels, significantly reduced serum insulin and C-peptide levels (p<0.05), and reduced HOMA-IR (p<0.001) after two weeks of treatment in ZDF rats.
- BMF-219 showed prolonged glycemic control as evidenced by decreased glucose levels during an oral glucose tolerance test on day 15 (AUC reduction of 54%, p<0.001) and on day 29 during the drug washout period (AUC reduction of 40%, p<0.05, ~2 weeks after the last dose) in the ZDF model, indicating durable glycemic control.
- Strikingly, BMF-219, but not pioglitazone, reduced blood glucose levels by OGTT in STZ animals (AUC reduction of 41%, p<0.05).
- Significant reductions in blood lipemic levels (p<0.01) and body weight were observed in both models.
- Collectively, our data indicate the novel and marked potential of BMF-219 as an oral, long-acting treatment for T2DM.

References

- Ma, J. et al. Menin-regulated Pbk controls high fat diet-induced compensatory beta cell proliferation. *EMBO Mol Med*. 2021;13(5):e13524.
- Yang, Y. et al. Deletion of the *Men1* gene prevents streptozotocin-induced hyperglycemia in mice. *Exp Diabetes Res.* 2010;2010:876701.

biomeafusion.com Biomea Fusion, Inc. / 900 Middlefield Road, 4th Floor / Redwood City, CA 94063

Acknowledgements – We acknowledge Charles River Laboratories for animal studies.